INFRADURE: SUSTAINED DELIVERY OF INTERFERON-ALPHA-2b IN SCID MICE BY IMPLANTED IFNα-2b SECRETING DERMAL BIOPUMPS

B. Stern[1], E. Shoshani[1], A. Pearlman[1], R. Meari[1], H. Goltzmann[1], A. Rimler[1], A. Okun[1], H. Bookay[1], T. Gibly[1], P. Ng[2], E. B. Keeffe[3] [1]Medgenics, Inc, Misgav, Israel; [2]Molecular Genetics, Baylor College of Medicine, Houston, TX, United States; and [3]Division of Gastroenterology and Hepatology, Department of Medicine, Stanford University School of Medicine, Stanford, California, United States

OVERVIEW

The Need: For a much more tolerable and cost effective interferon therapy

- **Even if DAA's work but cost significantly more than current standard of care, many patients will not be able to afford and many payers will not be prepared to pay for a marginal improvement**
- **There will thus remain a need for a better tolerated, more cost effective interferon therapy**

Potential Solution: This poster presents a novel solution – INFADURE: a tissue implant providing 6 months or more continuous delivery of IFNα from a single administration

Introduction and Rationale

Hepatitis C affects approximately 170 million people worldwide, yet only half of the patients respond to the current standard of care of PEGinterferon and ribavirin, which cause substantial adverse effects. Schenker et al (1) have shown that it may be possible to use continuous delivery of IFNα to achieve comparable HCV eradication with significantly reduced side effects while using lower levels of IFNα, as opposed to frequent high-dose bolus injections. To develop a practical way to implement such sustained low level IFNα therapy for use in patients, we are developing an application of Biopump: a platform technology using autologous 2mm x 30mm micro-organ tissue explants taken from the patient's dermis by needle biopsy, and processed ex-vivo in 10-14 days to provide sustained production and secretion of proteins (as explained below). We have reported proof of concept of such Biopumps producing and secreting sustained high levels of erythropoietin in vitro and have shown that following subcutaneous administration in the anemic patients from which they were prepared, have elevated their hemoglobin levels for 6-12 months or more (2). We have also reported at this conference (4) that the same Biopump platform has now been successfully adapted to make INFRADURE Biopumps which produce and secrete more than 1 µg /day of IFNα per Biopump for more than 4 months in vitro, which could provide continuous IFN therapy over a similar period of time in treatment of patients with chronic Hepatitis C. We now aim to demonstrate dose-dependent delivery of active IFNα by subcutaneous implantation of such human INFRADURE Biopumps into a SCID mice animal model.

Biopump Platform Technology: Introduction

An INFRADURE Biopump is produced from a unique excised elongated primary tissue explant called a micro-organ, typically measuring 30 mm in length and 1-2 mm in diameter. As shown in Figure 1, this tissue is harvested directly from the patient's dermis under local anesthetic, using of a proprietary device we have developed, the DermaVac. The harvest sites heal within 2-3 weeks, as seen in Figure 3 for a typical patient from our recent clinical trial. The micro-organ is intact tissue and can be viably maintained for weeks ex vivo, if desired, although only 10-14 days are required to modify them ex vivo to produce the protein-secreting Biopumps. Sustained action Biopumps are produced by ex vivo transduction of micro-organs with Helper Dependent Adenoviral vectors (HDAd) that encode the IFNα gene or the gene for any other desired therapeutic protein – Biopump is a platform technology for producing a wide range of proteins (see below). The resultant Biopumps express and secrete the therapeutic protein, and following measurement of daily production rate, the requisite dose is administered by subcutaneous implantation of the corresponding number of Biopumps, in order to provide continuous delivery of a known amount of the therapeutic protein in the patient. A schematic representation of the steps in Biopump preparation and administration to the patient is shown in Figure 1.

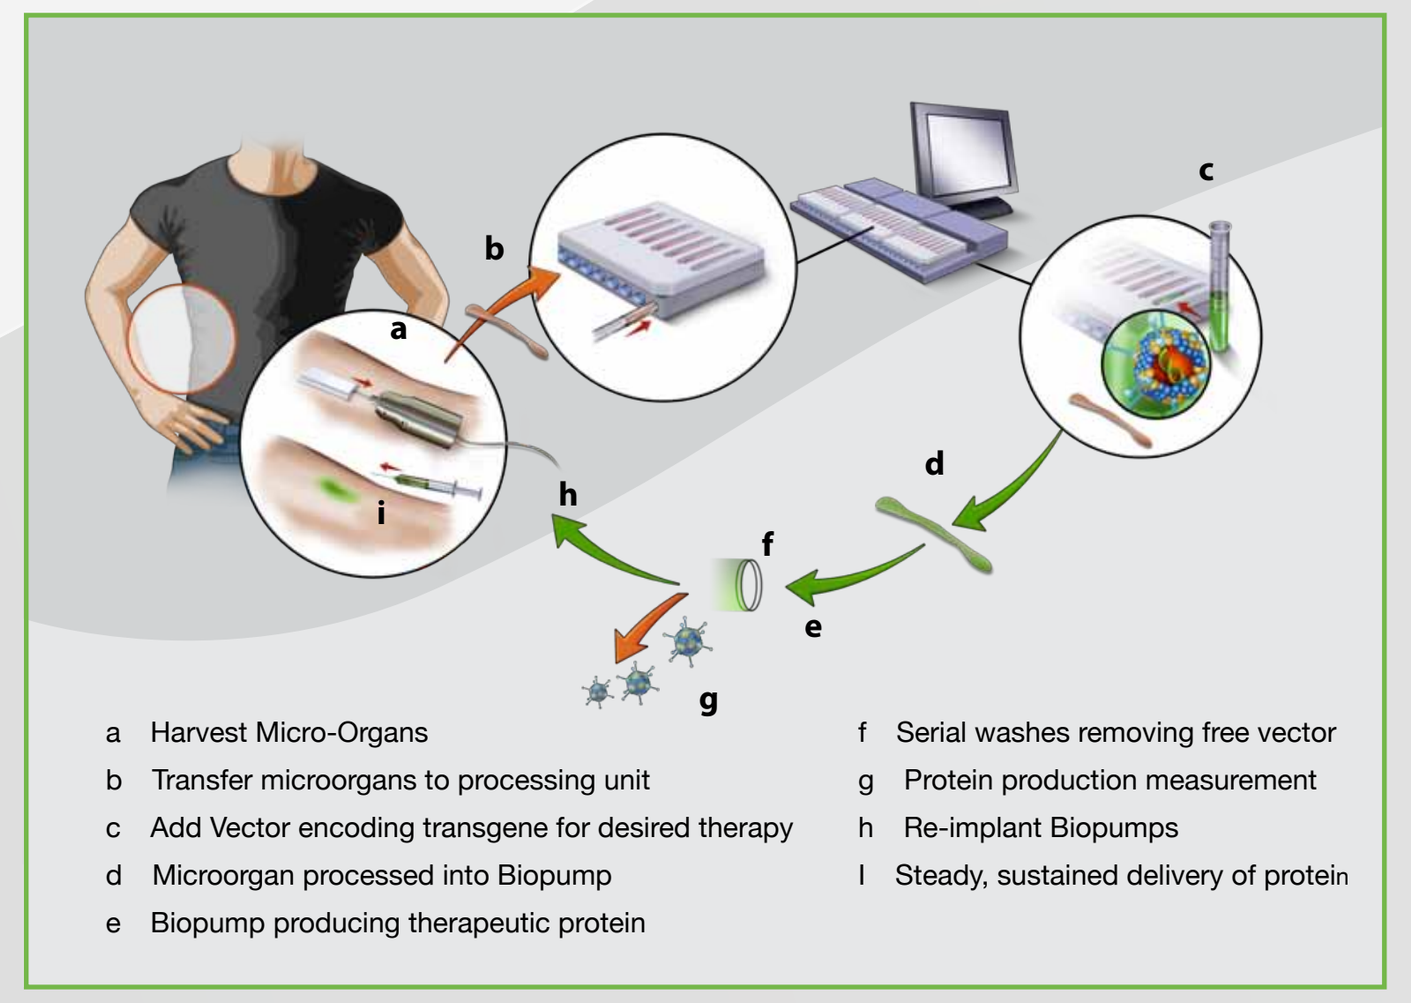
a Harvest Micro-Organs
b Transfer microorgans to processing unit
c Add Vector encoding transgene for desired therapy
d Microorgan processed into Biopump
e Biopump producing therapeutic protein
f Serial washes removing free vector
g Protein production measurement
h Re-implant Biopumps
I Steady, sustained delivery of protein

Figure 1 Biopump Preparation

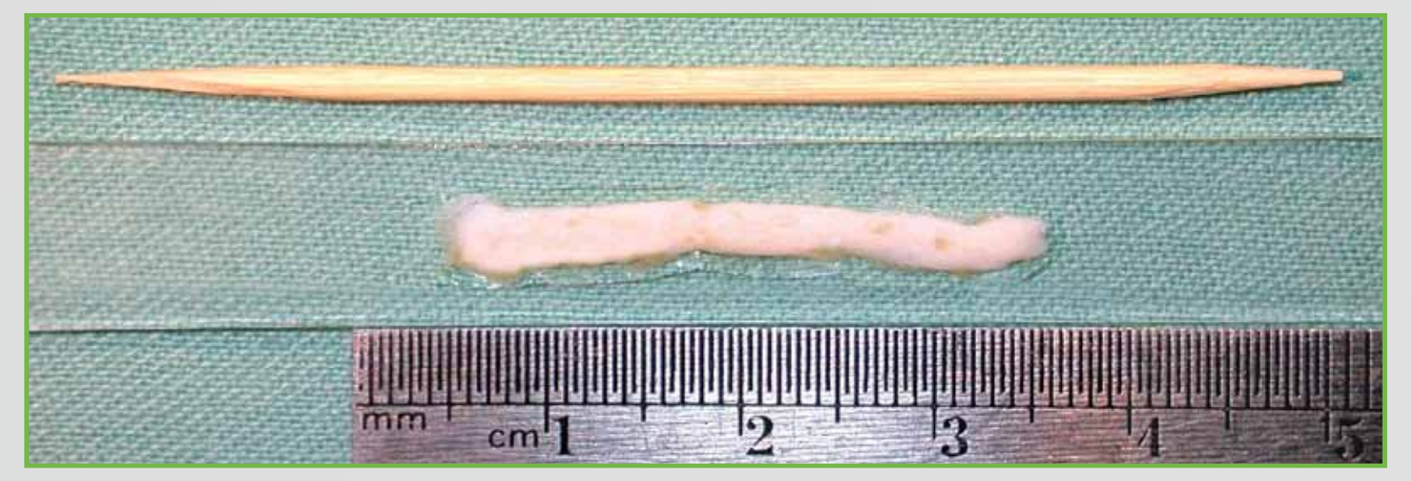

Figure 2 Typical Dermis Micro-Organ vs Toothpick

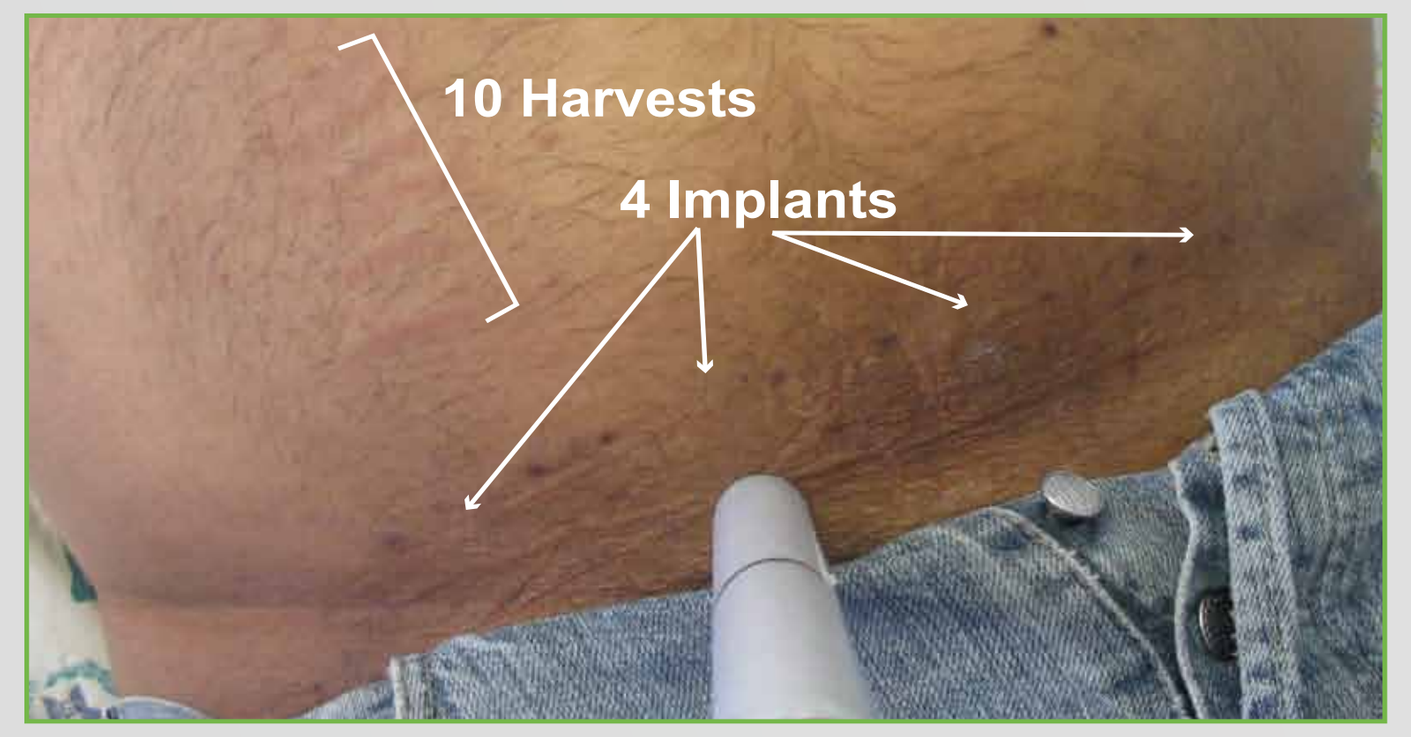
10 Harvests
4 Implants

Figure 3 Shown are the sites where 10 micro-organs were harvested, and where 4 EPODURE Biopumps were implanted, as demarcated by tattoo dots

Developing Potent INFRADURE Biopumps

Objectives

To test the dose-response, the duration of sustained delivery, and the biological activity, of IFNα as continuously produced and delivered by a single administration of human INFRADURE Biopumps in a SCID mouse model. A synopsis of experiments performed to address these is presented here.

Methods

Human skin source: As described in more detail elsewheree in this conference (4), excised skin from healthy individuals undergoing routine abdominal resection (tummy-tuck operations) was obtained under ethical approval. From these, human skin micro-organs were harvested, processed to become INFRADURE Biopumps producing hIFNα, and maintained for ten days in vitro before their use in animal tests.

Animal model: SCID mice: To test in vivo performance of INFRADURE Biopumps prepared from human skin, an animal model is required which will not reject the human dermis tissue. Severe Combined Immunodeficiency, or SCID, mice which cannot make T cells or B cells, are a suitable animal model and we have previously reported their use in conjunction with Biopumps producing another protein, erythropoietin, to demonstrate long term delivery of active protein into the mouse blood circulation (3). We have obtained the FDA's recommendation for using it for our toxicological studies. This study uses a similar approach but employs human dermis INFRADURE Biopumps whose daily production of hIFNα in vitro was measured one day prior to implantation into the mice. Each SCID mouse was implanted with two INFRADURE Biopumps which together secreted at least 1 µg/day of hIFNα, and blood samples were taken approximately every 10 days thereafter. Serum levels of hIFNα were measured by Verikine ELISA kit (PBL #41100), and biological activity of the hIFNα was determined using the iLite Human Interferon Alpha kit (PBL, Product #51100). The activity kit is a bioluminescence assay designed for the quantitative determination of IFNα activity in interferon-sensitive cells. The study was terminated 123 days after implantation, with necropsy performed on the mice, including samples from kidneys, liver, spleen and implantation site (implant and surrounding tissue).

Dose-dependent Delivery

To test dose response, Biopumps were implanted subcutaneously in SCID mice to deliver either a "lower" dose of 1300ng/day to each of 4 mice, or a "higher" dose of 4000 ng/day to each of 2 mice. Blood samples were taken every 10-14 days afterwards for 105 days. As see in Figure 4, serum hIFNα levels reflected the high implanted doses, appropriate for 70 Kg humans rather than 20g mice, and reached extraordinarily high levels on day 10: "low" group averaged 1700 pg/ml, "high" group averaged 8,900 pg/ml.

Sustained production and Delivery in SCID Mice

Figure 4 also demonstrates that INFRADURE Biopumps continue to secrete hIFNα in SCID mice for extended duration (over 100 days) from a single INFRADURE administration. The mice managed to reduce the initial overdose levels to standard therapeutic levels in the10 pg/ml range within 3-4 months, with serum levels of "higher" dose always greater than those of "lower" dose. The steady decline over weeks of the circulating hIFNα, could reflect mechanisms of the mice to avoid toxicity due to administration of a dose for 70 Kg in a mouse weighing 20g. It is noted that the necropsy study showed no signs of toxicity after 123 days' continuous treatment. We have shown elsewhere (3) that implantation of EPODURE Biopumps producing human-therapeutic doses of erythropoietin in SCID mice likewise shows rapid decline to mouse-therapeutic levels in the mouse serum, whereas implantation of similar dose in treating anemic patients has resulted in sustained elevation of their hemoglobin levels for 6-12 months and more from a single administration (2).

hIFNα Bio-activity

In a separate experiment, 12 SCID mice were implanted with INFRADURE biopumps secreting at least 1 µg/day. Blood samples were taken prior to implantation and ten days post-implantation, with hIFNα serum levels and their corresponding serum hIFNα biological activity measured, as shown in Figure 5A. All twelve mice showed zero hIFNα in their serum (both by activity and by ELISA) prior to implantation, and similar very substantial levels of hIFNα on both activity and ELISA on day 10. Correlation between hIFNα by ELISA versus corresponding hIFNα bio-activity on the same samples is shown in Figure 5B, where each dot represents a single mouse. Pre-implantation serum IFNα levels and activities were 0, and on day 10 serum IFNα ranged from about 3,000 to 6,000 pg/ml, correlated well to activity ranging from about 1300 to 2,500, with $R^2 = 0.965$. Y/X ratio was 2.28 pg/IU, indicating potency compares favorably to the 3.85 pg/IU of Intron-A per its package insert. These results confirm that INFRADURE is secreting potent hIFNα into the serum of these mice.

Biopump Platform Technology: Proof of Concept

The Biopump technology is capable of producing and delivering a wide range of proteins. To demonstrate the Biopump platform, Biopumps secreting different proteins have been prepared from micro-organs of the same skin sample.

In Figure 6, two sets of Biopumps were prepared from the same skin sample under comparable conditions to produce either IFNα or EPO. Each set produced sustained therapeutic levels in vitro of IFNα or EPO respectively. These results demonstrate the ability of the same Biopump platform technology to produce sustained high levels of different therapeutic proteins.

Dose-dependent delivery of hIFNα in SCID MICE

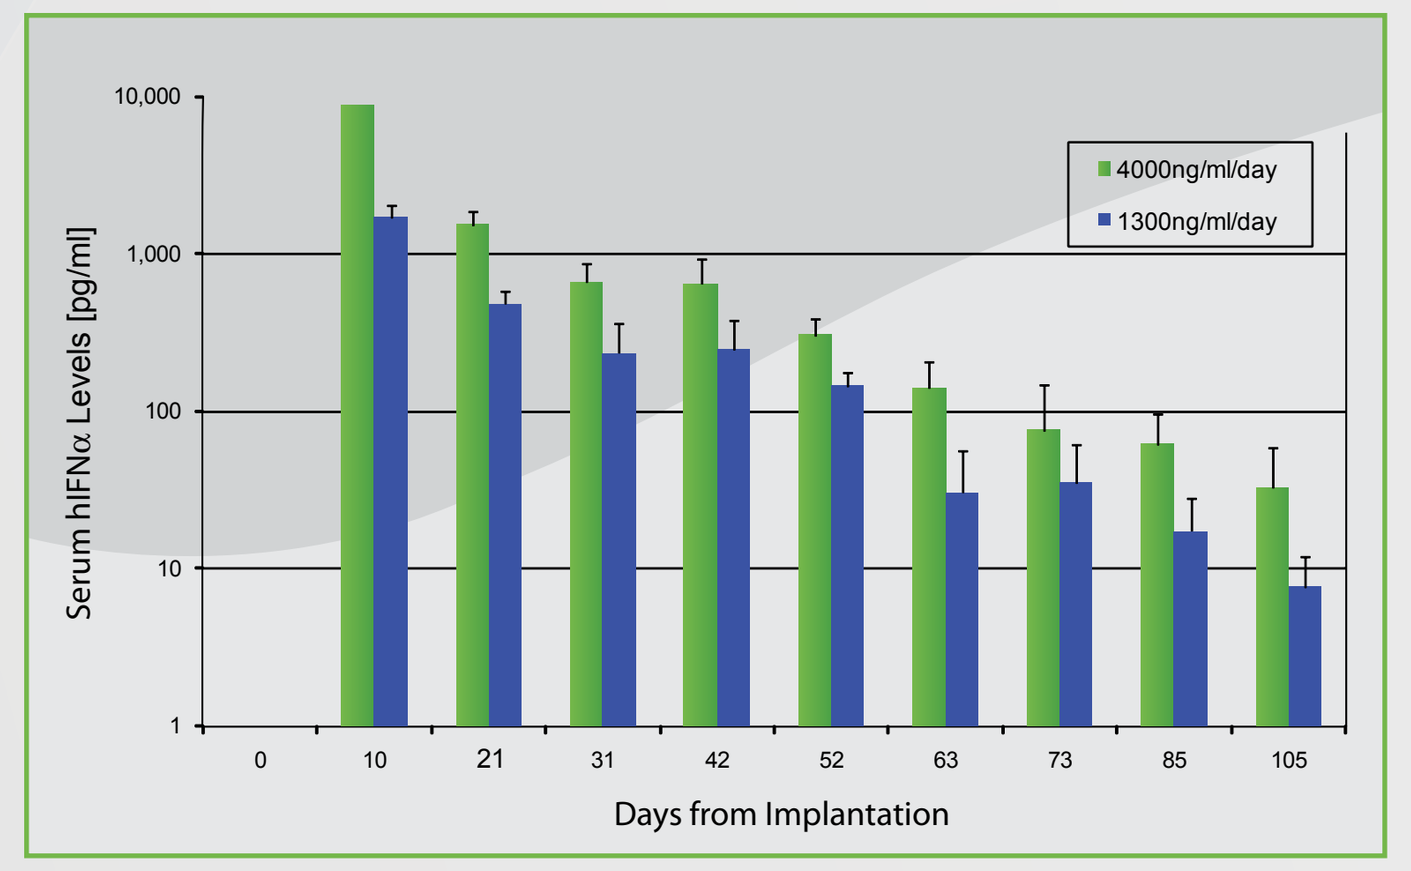
4000ng/ml/day
1300ng/ml/day
Serum hIFNα Levels [pg/ml]
Days from Implantation

Figure 4 Dose-dependent delivery of hIFNα in SCID mice implanted with INFRADURE Biopumps producing either 1300 ng/day (blue bars) or 4000 ng/day (red bars), as measured pre-implantation by ELISA.

Delivery of active IFNα in SCID mice

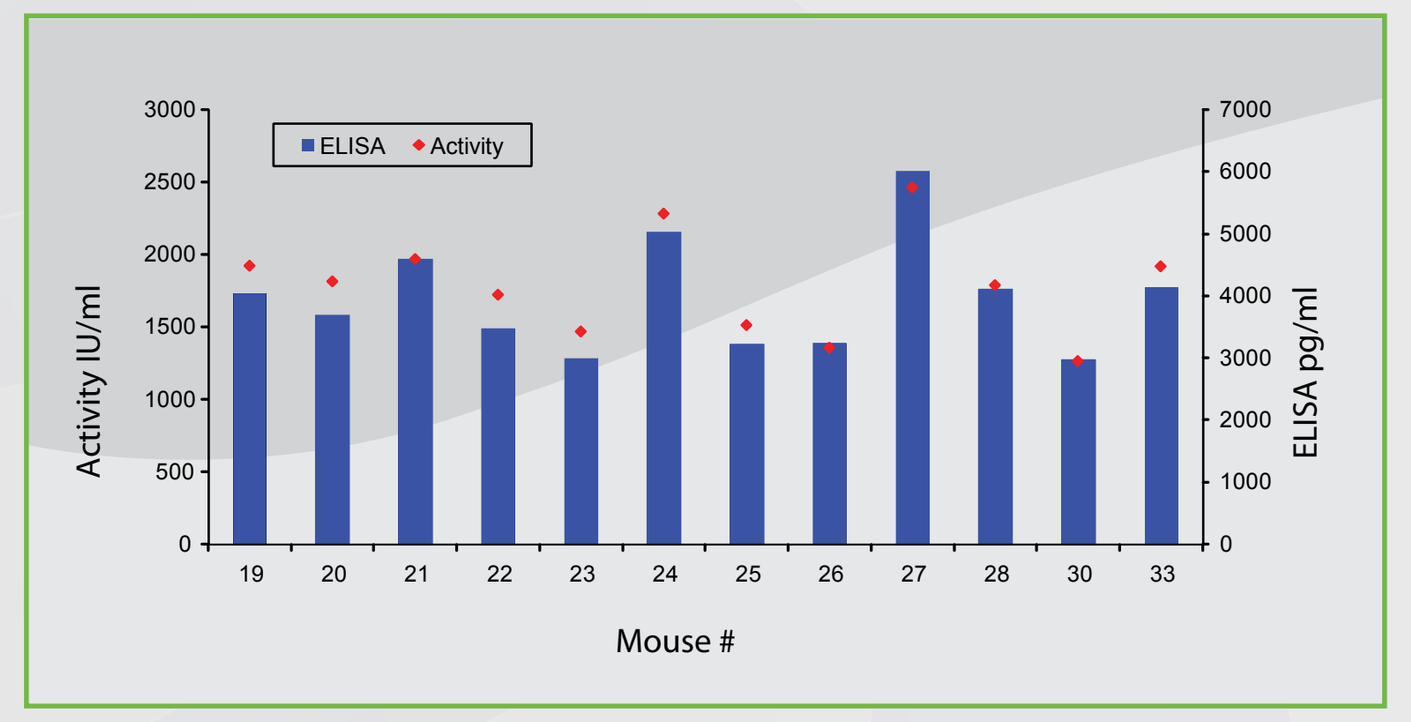
ELISA
Activity
Activity IU/ml
ELISA pg/ml
Mouse #

Figure 5A hIFNα serum levels in 12 mice implanted with INFRADURE and their corresponding Bio-activity. hIFNα levels and activity were measured 10 days post implantation. Blue bars correspond to hIFNα levels measured by ELISA (pg/ml), red diamonds correspond to hIFNα bio-activity measured by (IU/ml).

Correlation of IFNα activity and concentration in mouse serum

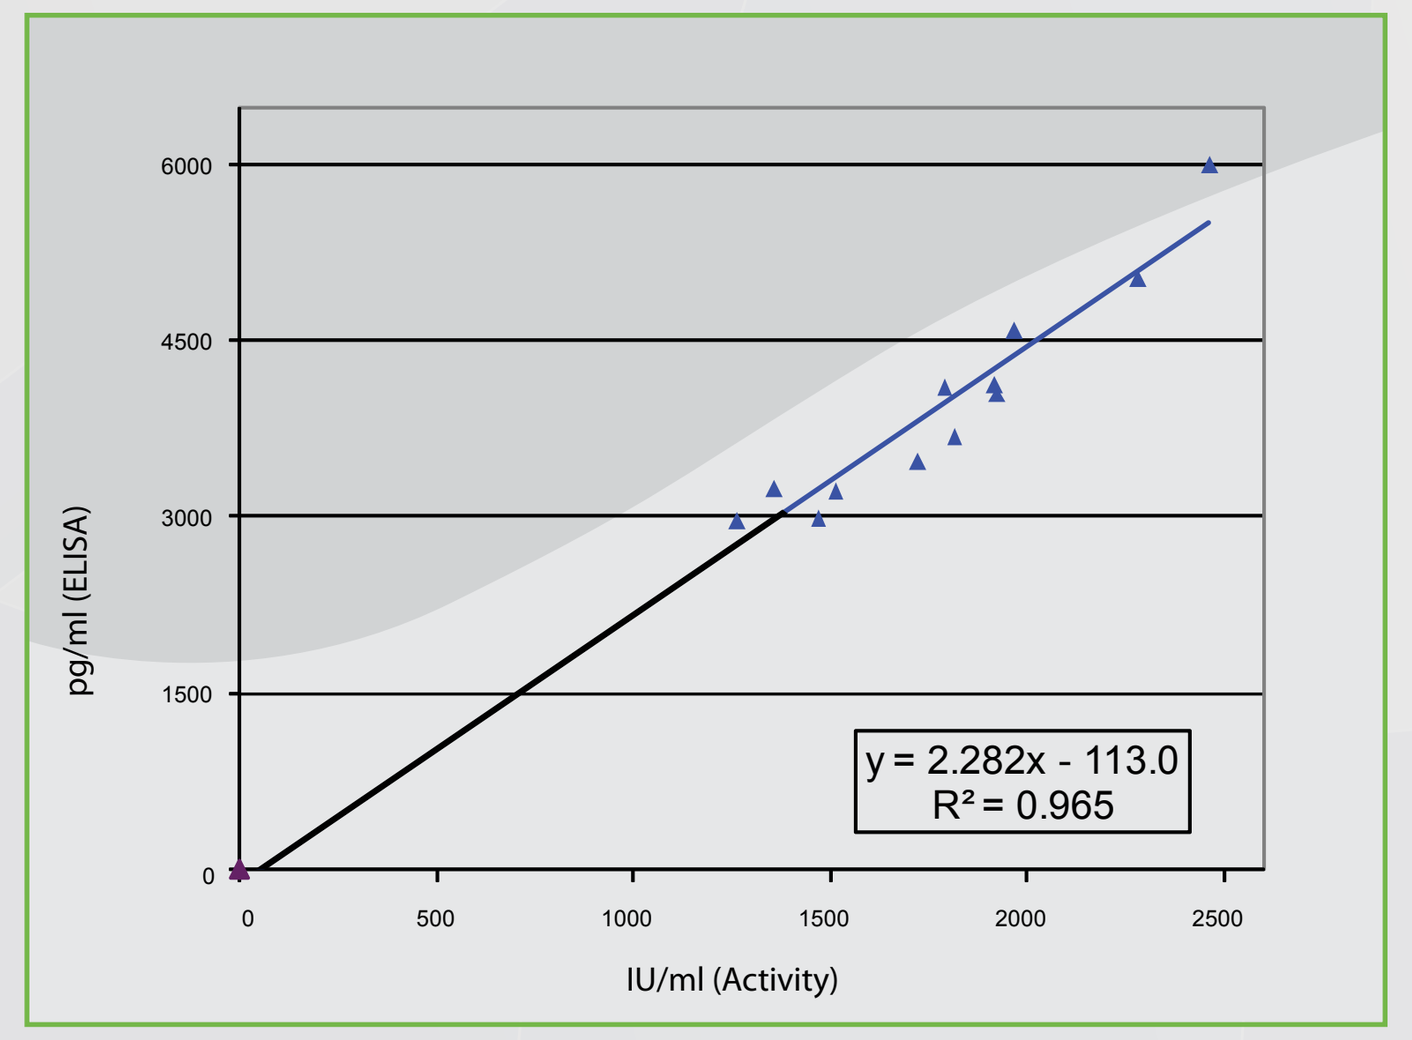
y = 2.282x - 113.0
R² = 0.965
pg/ml (ELISA)
IU/ml (Activity)

Figure 5 B Correlation between the hIFNα levels measured in the serum of INFRADURE implanted mice to their corresponding bio-activity.

Discussion

- Elsewhere in this conference (4) we have presented INFRADURE data showing the attainment of sustained levels of > 1 µg/day IFNα for more than 4 months (and counting) in vitro. By comparison with weekly dosing of Intron-A per its package insert, implantation of a few such INFRADURE Biopumps in a patient should provide sustained IFNα therapy.
- The results here show that subcutaneous implantation of INFRADURE Biopumps producing human-therapeutic doses of hIFNα in SCID mice delivers the hIFNα into the serum in a dose-dependent manner for more than 4 months, without signs of toxicity.
- The results further show that biological activity of hIFNα delivered in the mouse serum correlates to serum hIFNα concentration as measured by ELISA, with potency comparing favorably to that of Intron-A.
- We have elsewhere reported single administration of EPODURE Biopumps in anemic patients (2) and this has stabilized and now sustained elevated hemoglobin levels for 6-12 months and more.
- Hence, these INFRADURE results in SCID mice further support the suggestion that subcutaneous implantation of INFRADURE Biopumps in patients should deliver steady target levels of IFNα to help in the treatment of chronic Hepatitis C.
- Such continuous delivery could help reduce HCV RNA on a sustained basis for 6-12 months from a single administration, while significantly attenuating the side effects associated with bolus IFNα injections.
- INFRADURE thus represents a potentially superior, long lasting interferon therapy which is a viable alternative to bolus injections and to the use of mechanical infusion devices requiring refill of protein.
- This study supports the plan to proceed to clinical studies of INFRADURE in the treatment of Hepatitis C.

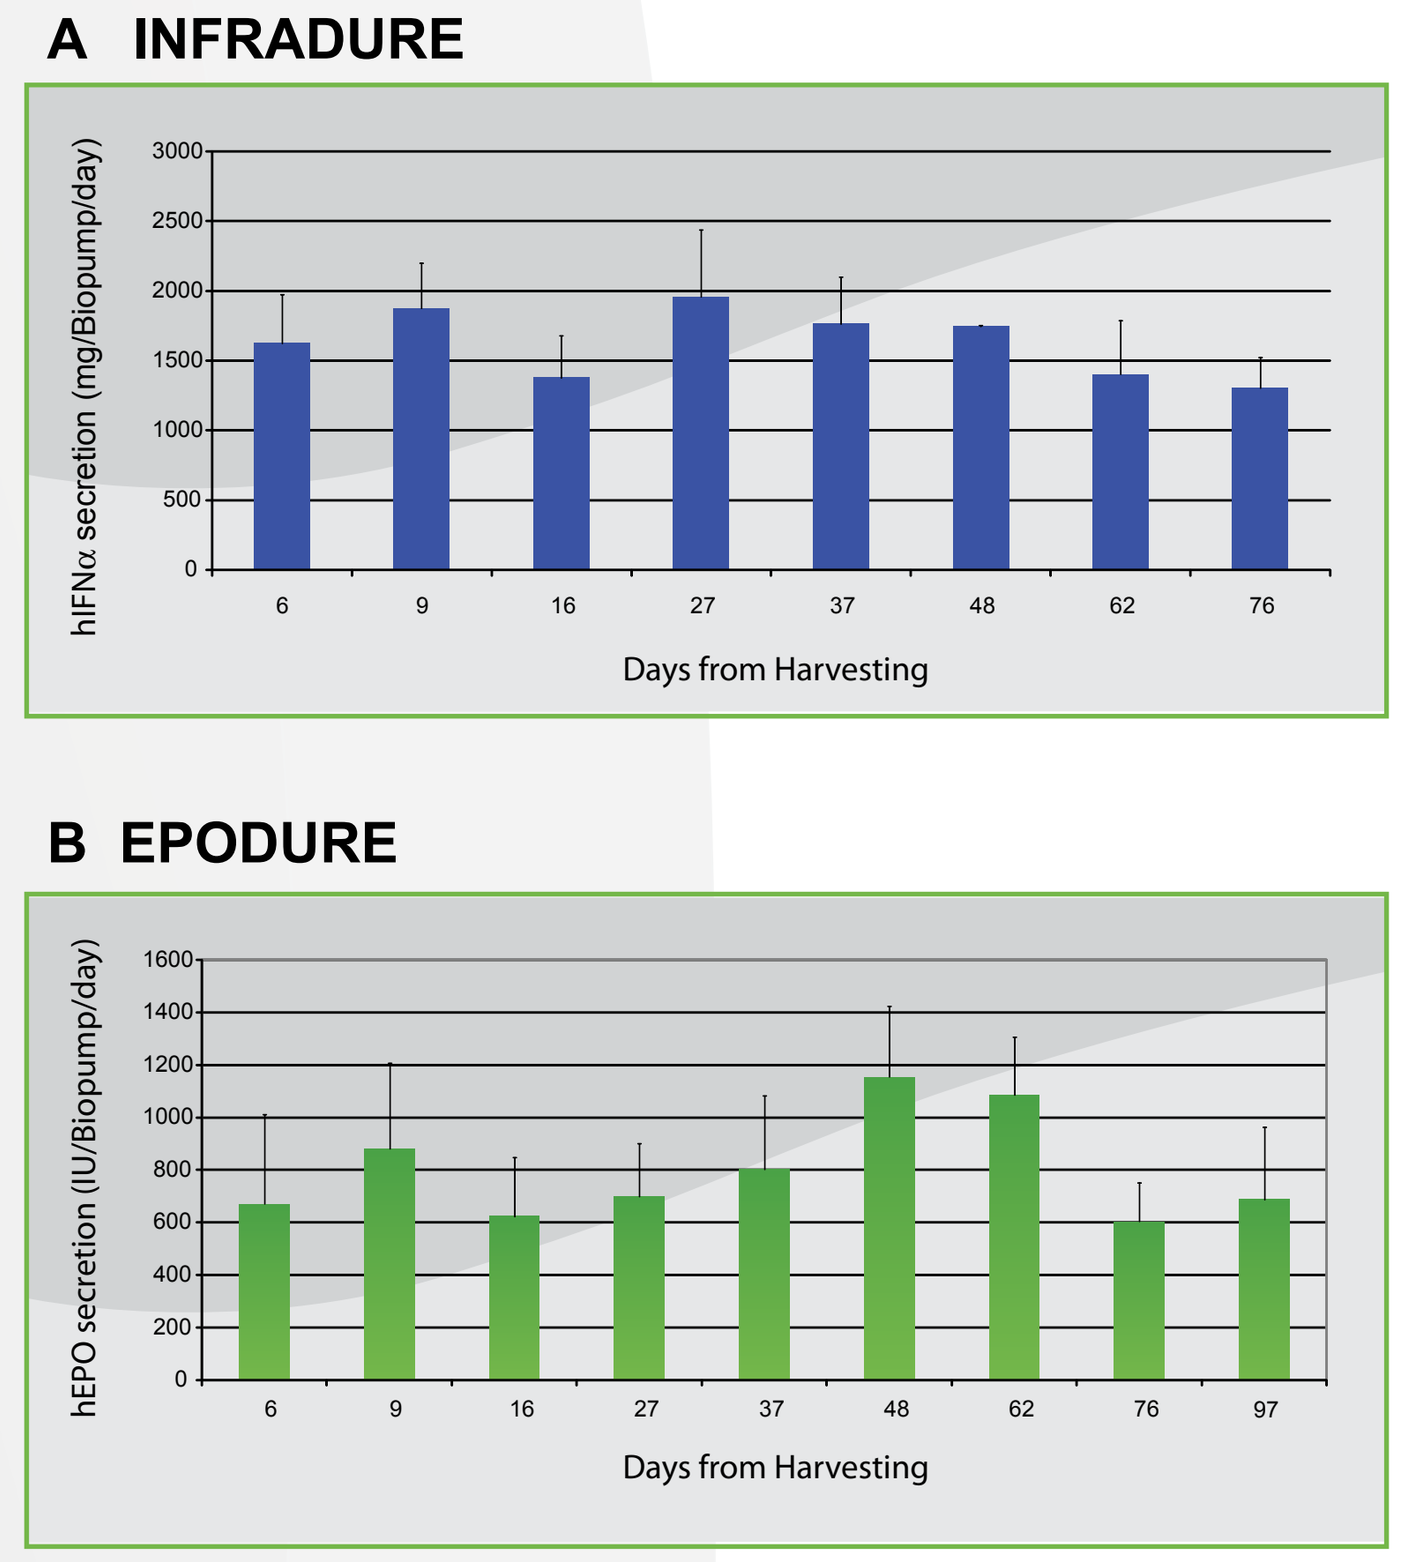
A INFRADURE
hIFNα secretion (mg/Biopump/day)
Days from Harvesting
B EPODURE
hEPO secretion (IU/Biopump/day)
Days from Harvesting

Figure 6 Protein production of Biopumps prepared from the same skin sample and processed to produce either hIFNa (Panel A blue bars) or hEPO (Panel B green bars): protein levels were measured by specific ELISA kits for the respective proteins.

Towards Practical, Cost-Effective Implementation

- For practical scale-up to cost-effective implementation, we are developing automated processing stations utilizing sealed cassettes to process Biopumps in a sterile, reliable manner.
- INFRADURE Biopumps are robust and enable practical transport from harvest by local physicians to processing at a centralized or regional facility, and back to local treatment sites for administration.
- As a result, we believe INFRADURE will be able to offer a more cost effective interferon therapy than today's standard of care.

Conclusions

INFRADURE Dose-dependent delivery
Delivery of hIFNα in SCID mice has been shown to correlate with administered INFRADURE dose

INFRADURE sustained delivery
Sustained delivery from a single administration has been reproducibly shown for more than 4 months in SCID mice, without toxicity

INFRADURE Potency
Biological activity of delivered hIFNα correlates with serum IFNα concentration, with potency comparing favorably to published values for commercial IFNα

Biopump Platform Demonstrated
Biopumps producing different proteins in micro-organs taken from the same skin sample prove that Biopumps are a platform technology

Potentially superior, cost-effective IFNα treatment for chronic Hepatitis C
Single administration could provide cost effective treatment with IFNα for 6-12 months with a much reduced side effect profile

REFERENCES

1. Schenker St, Cutler D, Finch J, Tamburro CH, Affirme M, Sabo R, Bay, M: Activity and tolerance of a continuous subcutaneous infusion of - interferon – α2b in patients with chronic Hepatitis C. J Interferon and Cytokine Res. 17: 665-670, 1997.
2. ASN 2008 abstract 552470
3. ASN 2008 abstract 552197
4. EASL 2010 abstract A-248-0025-00726

Israel
Teradion Business Park
P.O. Box 14
Misgav 20179 Israel

Phone: +972.4.902.8900
Fax: +972.4.999.0114

www.medgenics.com

U.S.A.
8000 Towers Crescent Dr.
Suite 1300
Vienna, Va. 22182 U.S.A.

Phone: 646.239.1690
Fax: 703.288.0070

info@medgenics.com